Facilitating Collective Action to Face the Opioid Overdose Crisis in Massachusetts

In 2019, RIZE launched Together in Recovery to foster an accessible, integrated treatment and recovery network in Massachusetts that champions evidence-based approaches, supports multiple pathways to recovery, and puts people in charge of their treatment choices.

rize massachusetts

Learning from Lived Experience: Understanding Barriers to Recovery

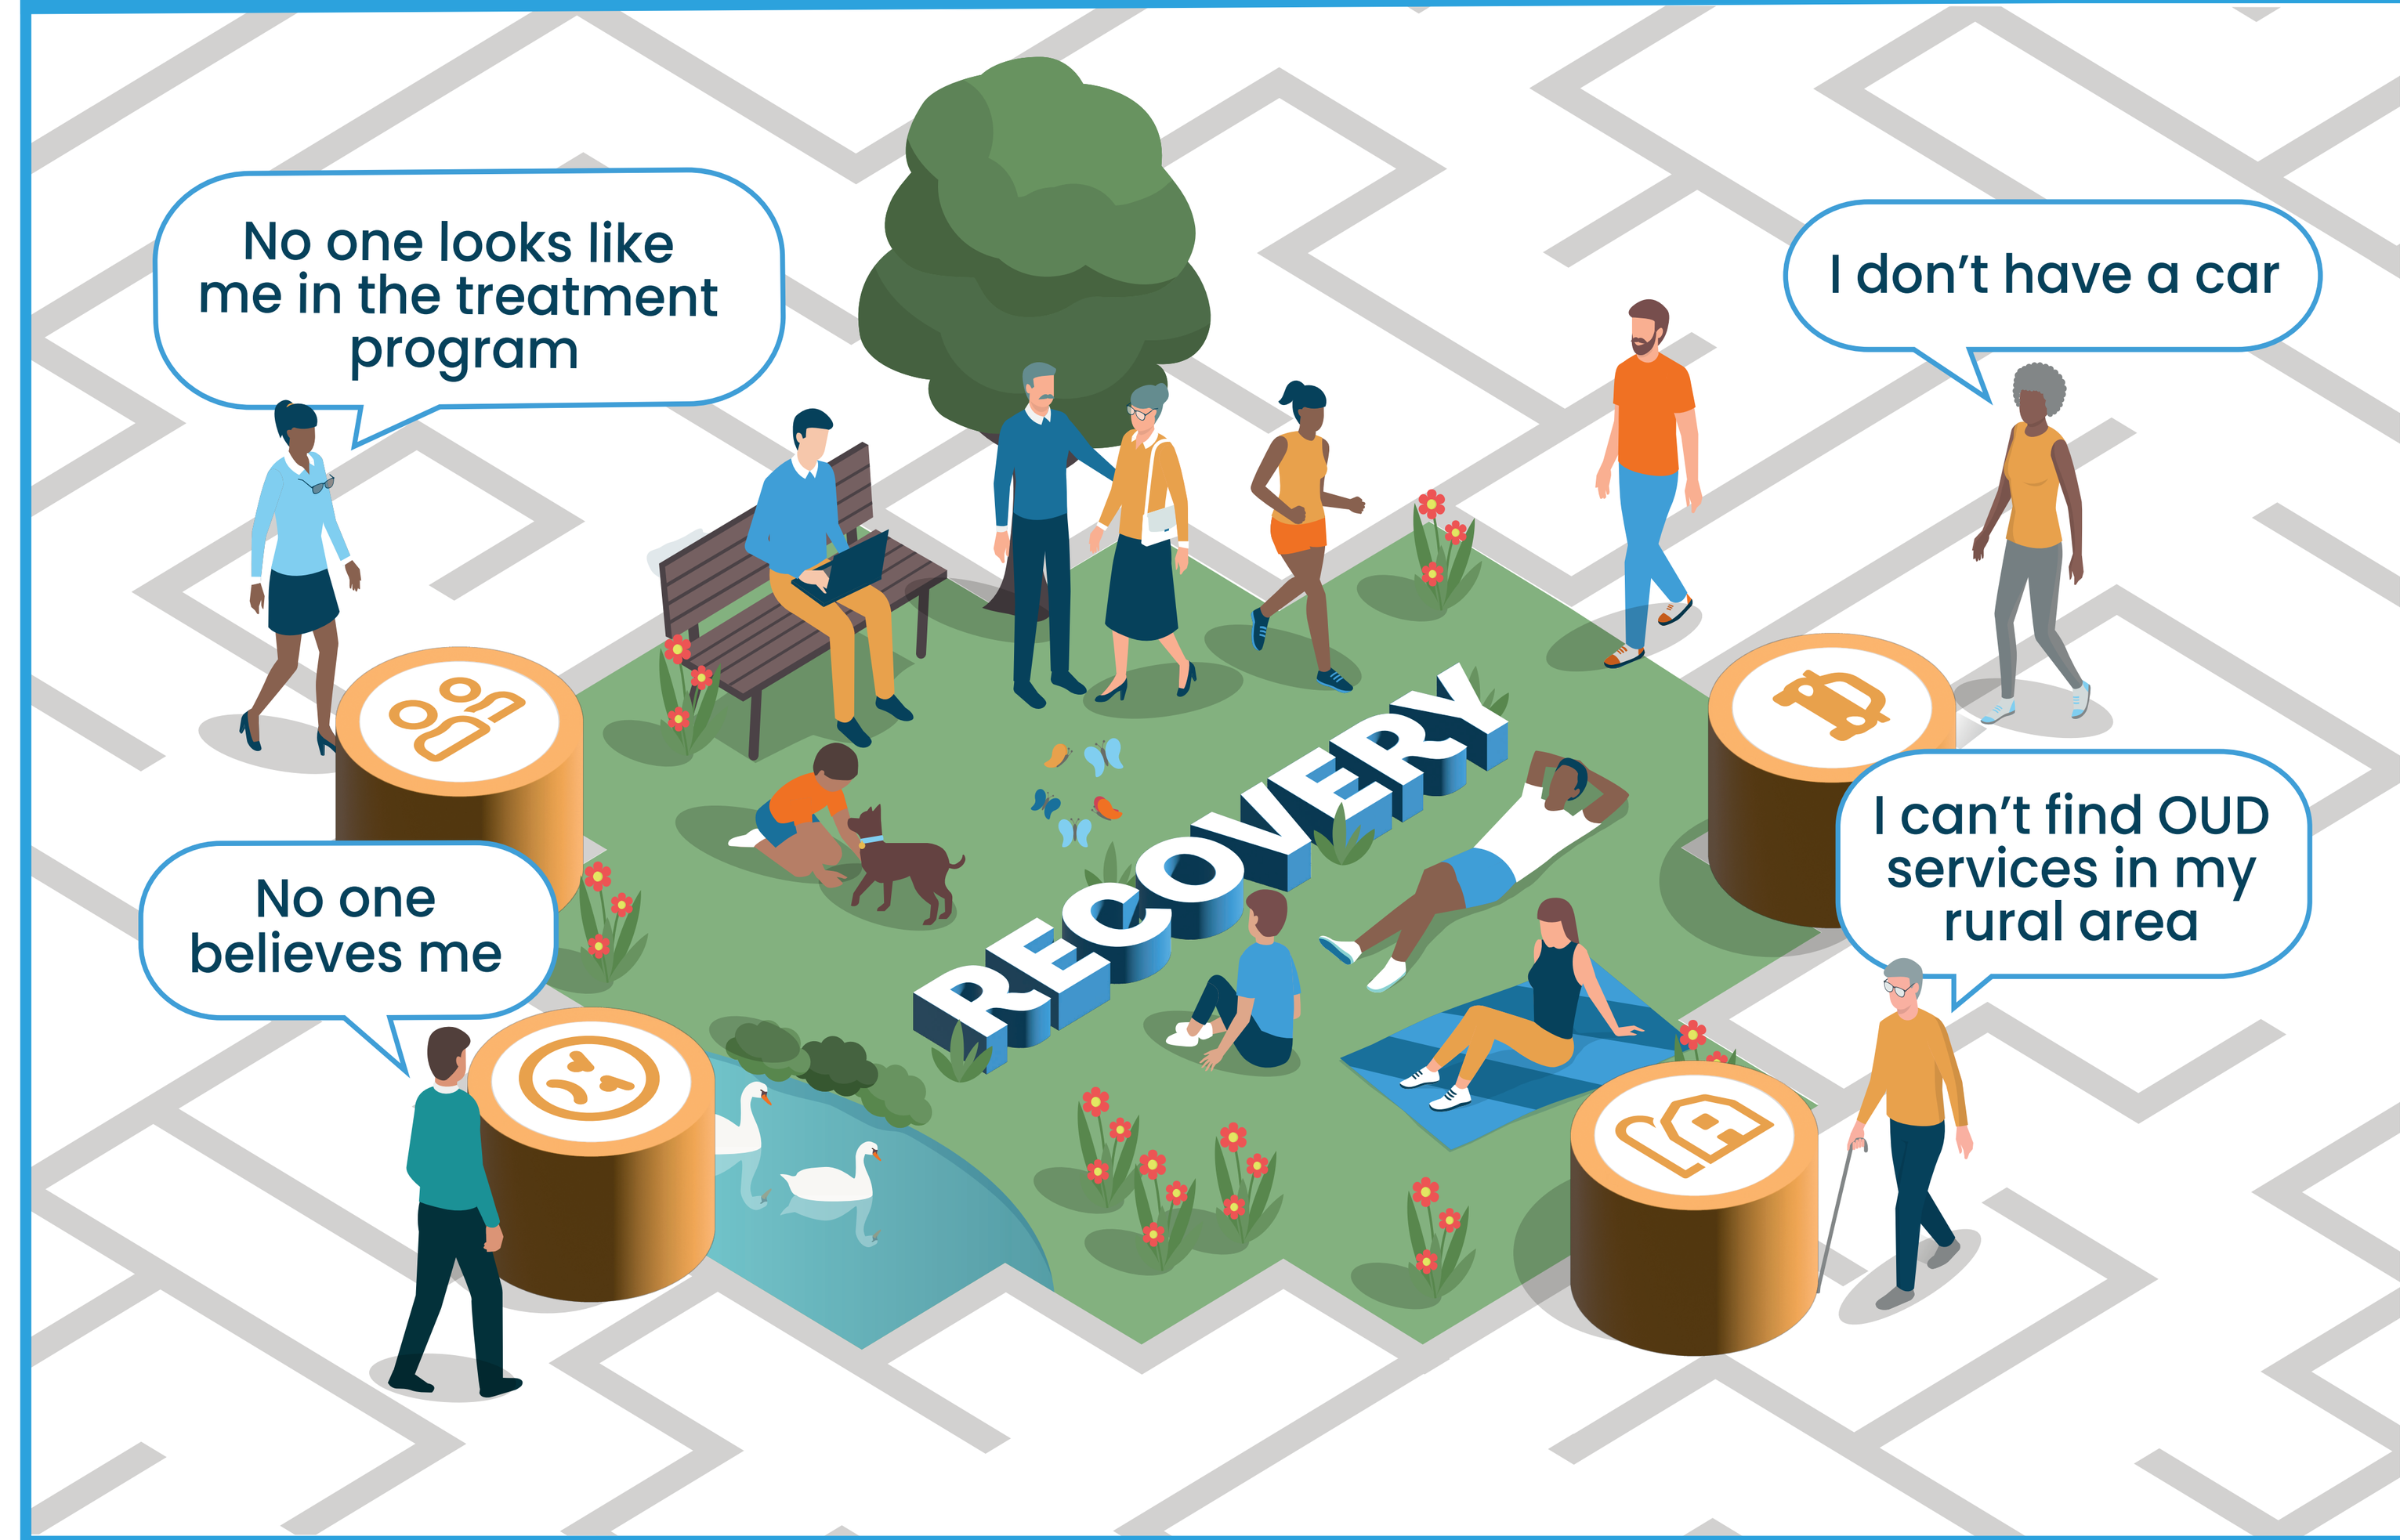
No one looks like me in the treatment program
I don't have a car
No one believes me
I can't find OUD services in my rural area
RECOVERY

Background

Despite the urgency of the opioid overdose crisis, long-standing divisions about how to support people with OUD endure. To disrupt this, we brought together a Change Team of 44 diverse stakeholders spanning peer, clinical, policy, family, and community roles, to create TiR. Guided by experienced facilitators, it convened over 300 people working to end the overdose crisis to exchange diverse viewpoints, build a shared vision, and identify collective action to ensure that a full range of treatment and recovery options are available to all.

Importance of Facilitated Dialogue

TiR engaged in intentional, and sometimes difficult, conversations to understand how unique perspectives and experiences shape convictions about the "right" pathways to recovery. TiR laid the foundation for what has become a lasting coalition evolving to meet the needs of the changing epidemic.

What We Learned

TiR created an unprecedented opportunity to bring people together to discuss what individuals, families, communities, and providers experience as they confront the opioid overdose crisis. TiR celebrated many accomplishments, including:

- Centering lived experience.
- Increasing knowledge and understanding of multiple pathways.
- Breaking down silos.
- Establishing common ground.

Through the process, TiR identified five top priorities for action:

1. Strengthening key transition points, identifying treatment gaps, and expanding the continuum of care to ensure longer-term support.
2. Enhancing workforce development and provider education.
3. Informing and influencing policy.
4. Building awareness of the roles of different agencies and expanding collaborations to increase access and avoid duplication.
5. Strengthening community-based organizations as an integral part of recovery.

Where We Are Headed

TiR will continue to support the high level of engagement and impact of its network, focusing on membership expansion, activation, and long-term agility to meet the changing nature of the epidemic. To address its priorities for action, TiR will embark on a new phase of work in 2022 to strengthen and uplift community-based organizations that meet the needs of residents in ways that are culturally responsive. Too often, these organizations are excluded from funding opportunities and decision-making networks, exacerbating racial and other inequities that drive disparities in health care. TiR aims to rectify these challenges by providing meaningful organizational development opportunities to address equity and improve care for people with substance use disorder.

By the Numbers

322 Number of regional dialogue meeting attendees

44 Change Makers

8 Regional meetings across the State

1 Statewide convening

MA

5 Top priorities identified

8 Priorities for Phase 2 established

1 Statement of principles developed

Quotes

After one particularly challenging conversation, a participant reflected that "it is hard to hate up close."

—TiR participant

. . . hearing many perspectives allowed me to examine my own mental models, and expanded my understanding of the barriers we are facing as a state to solve the issue of substance use disorder.

—Sydney Durand, The Phoenix and Change Team member

The regional meetings across the state had never been done and they helped us all to see the challenges that are being faced throughout every population.

—Tori Cyrus, Everett Fire Department and Change Team member

I appreciated learning from experts in real time at each meeting. Lastly, it was valuable to be able to call Together in Recovery colleagues for help and resources when we transitioned to working from home as a result of COVID-19.

—J. J. Bartlett, Fishing Partnership Support Services and Change Team member

We work in silos, but we don't realize it.

—Regional Dialogue Meeting participant

It is important for people from all disciplines to get involved and be part of the conversation to move forward.

—John McGahan, Gavin Foundation and Change Team member

SCAN ME

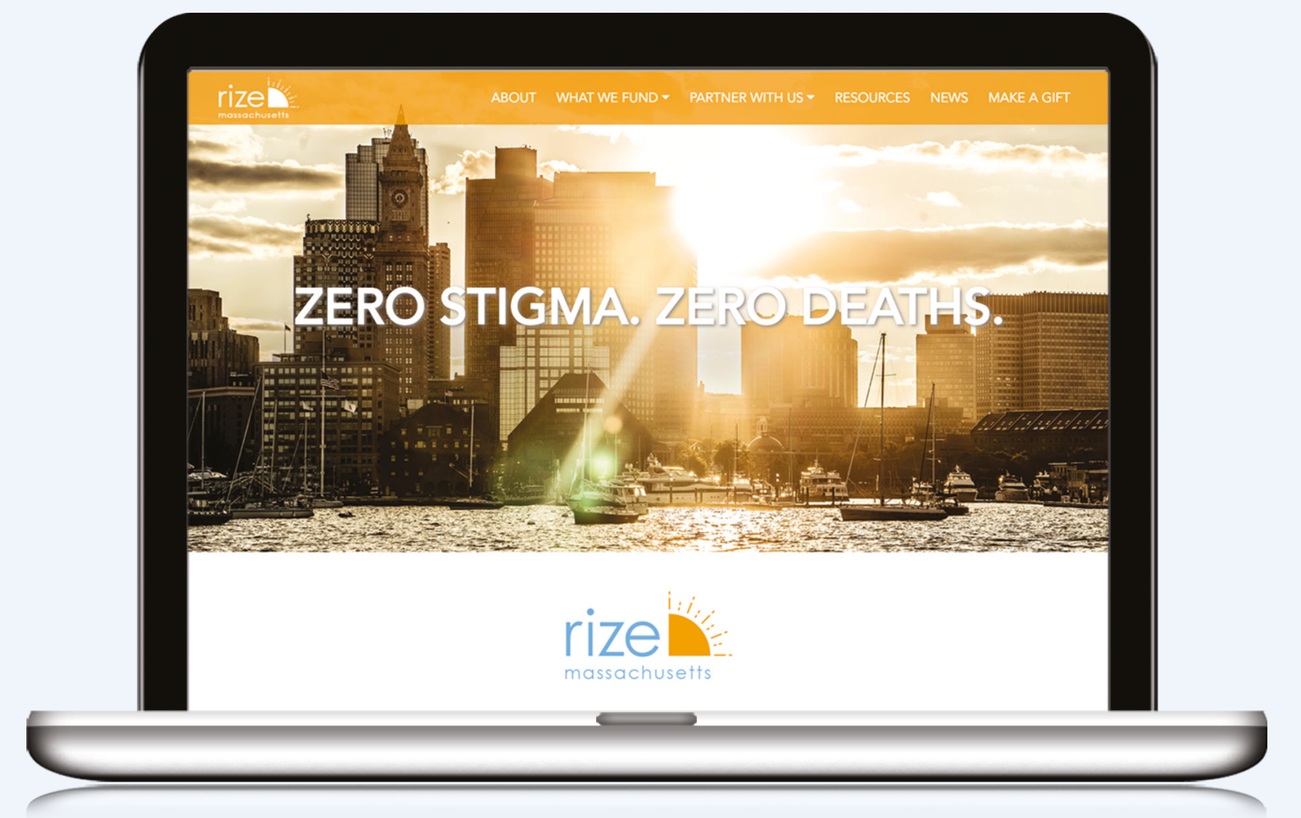
ZERO STIGMA. ZERO DEATHS.
rize

Materials

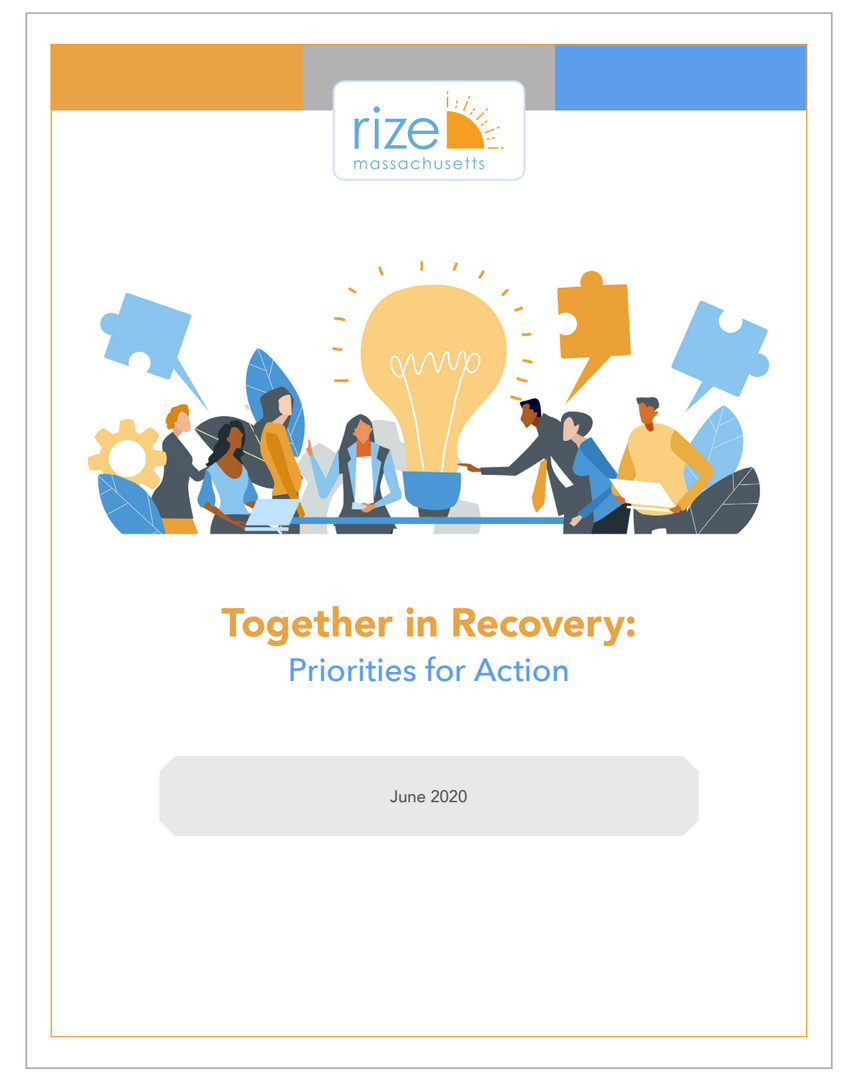
rize massachusetts
Together in Recovery:
Priorities for Action
June 2020

Priorities for Action

SCAN ME

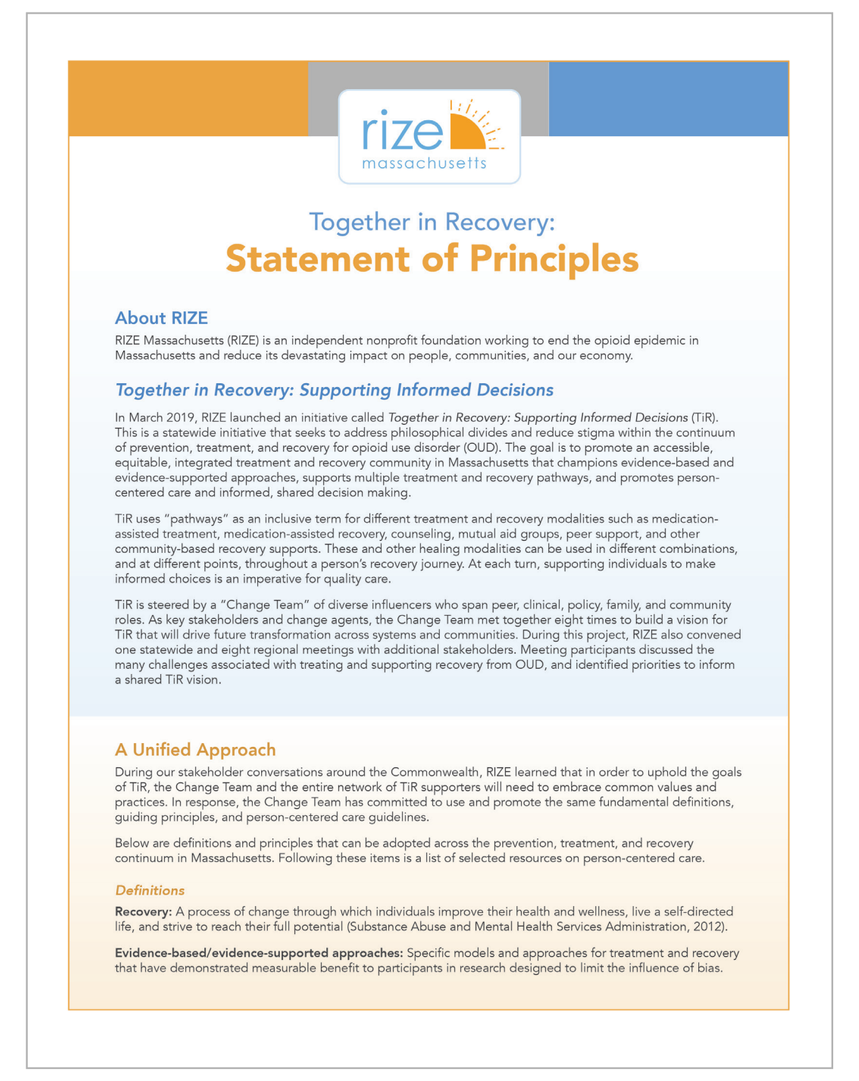
rize massachusetts
Together in Recovery:
Statement of Principles

Statement of Principles

SCAN ME

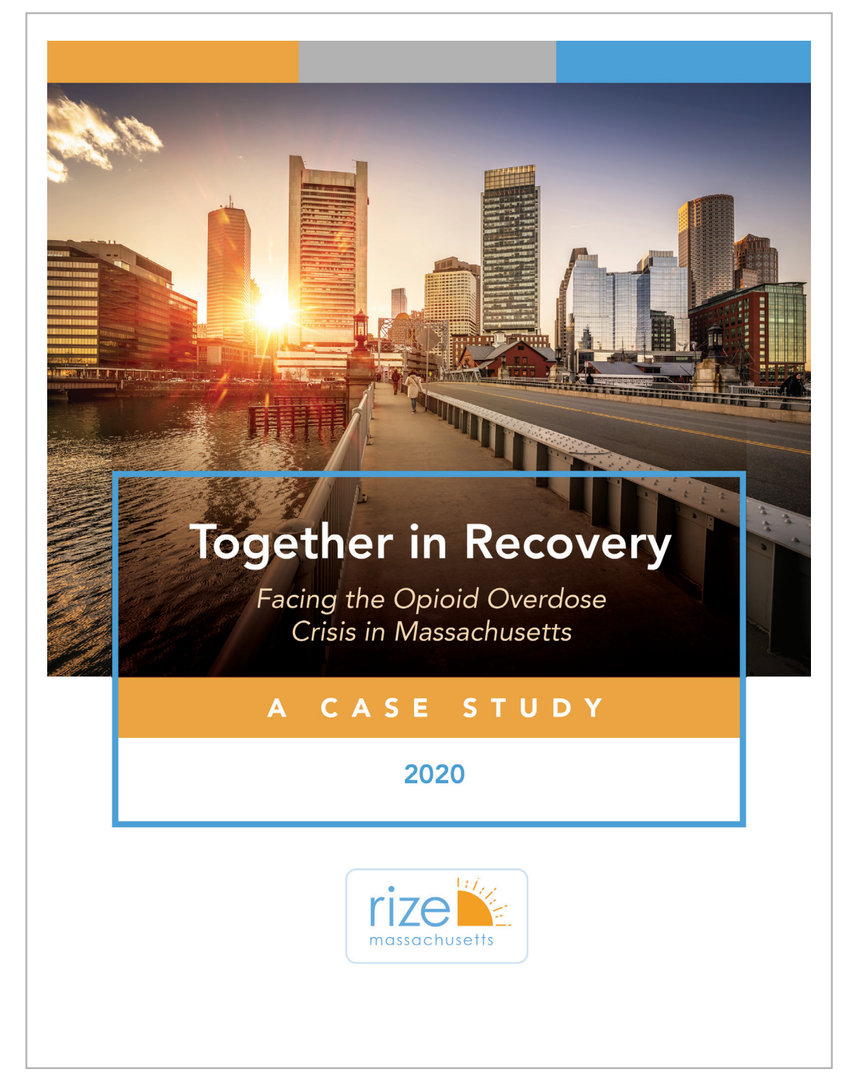
Together in Recovery
Facing the Opioid Overdose Crisis in Massachusetts
A CASE STUDY
2020
rize massachusetts

Case Study

SCAN ME

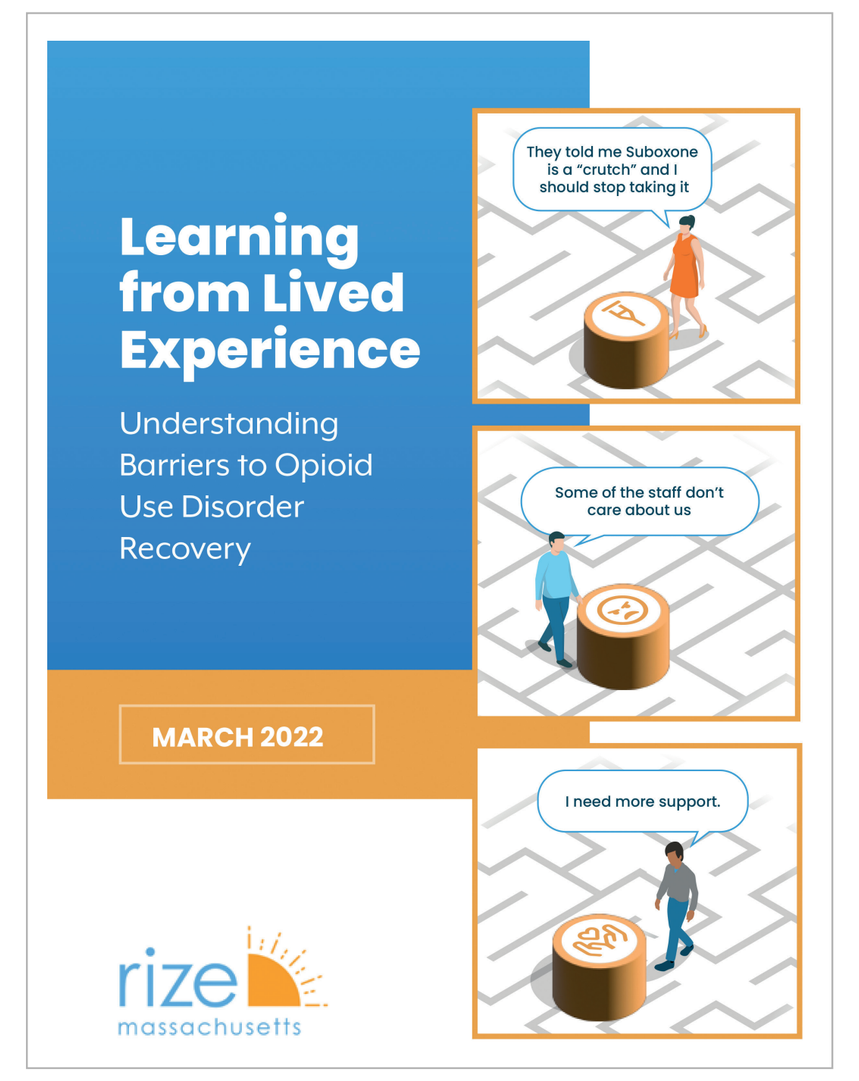
Learning from Lived Experience
Understanding Barriers to Opioid Use Disorder Recovery
MARCH 2022
rize massachusetts

Learning from Lived Experience

SCAN ME

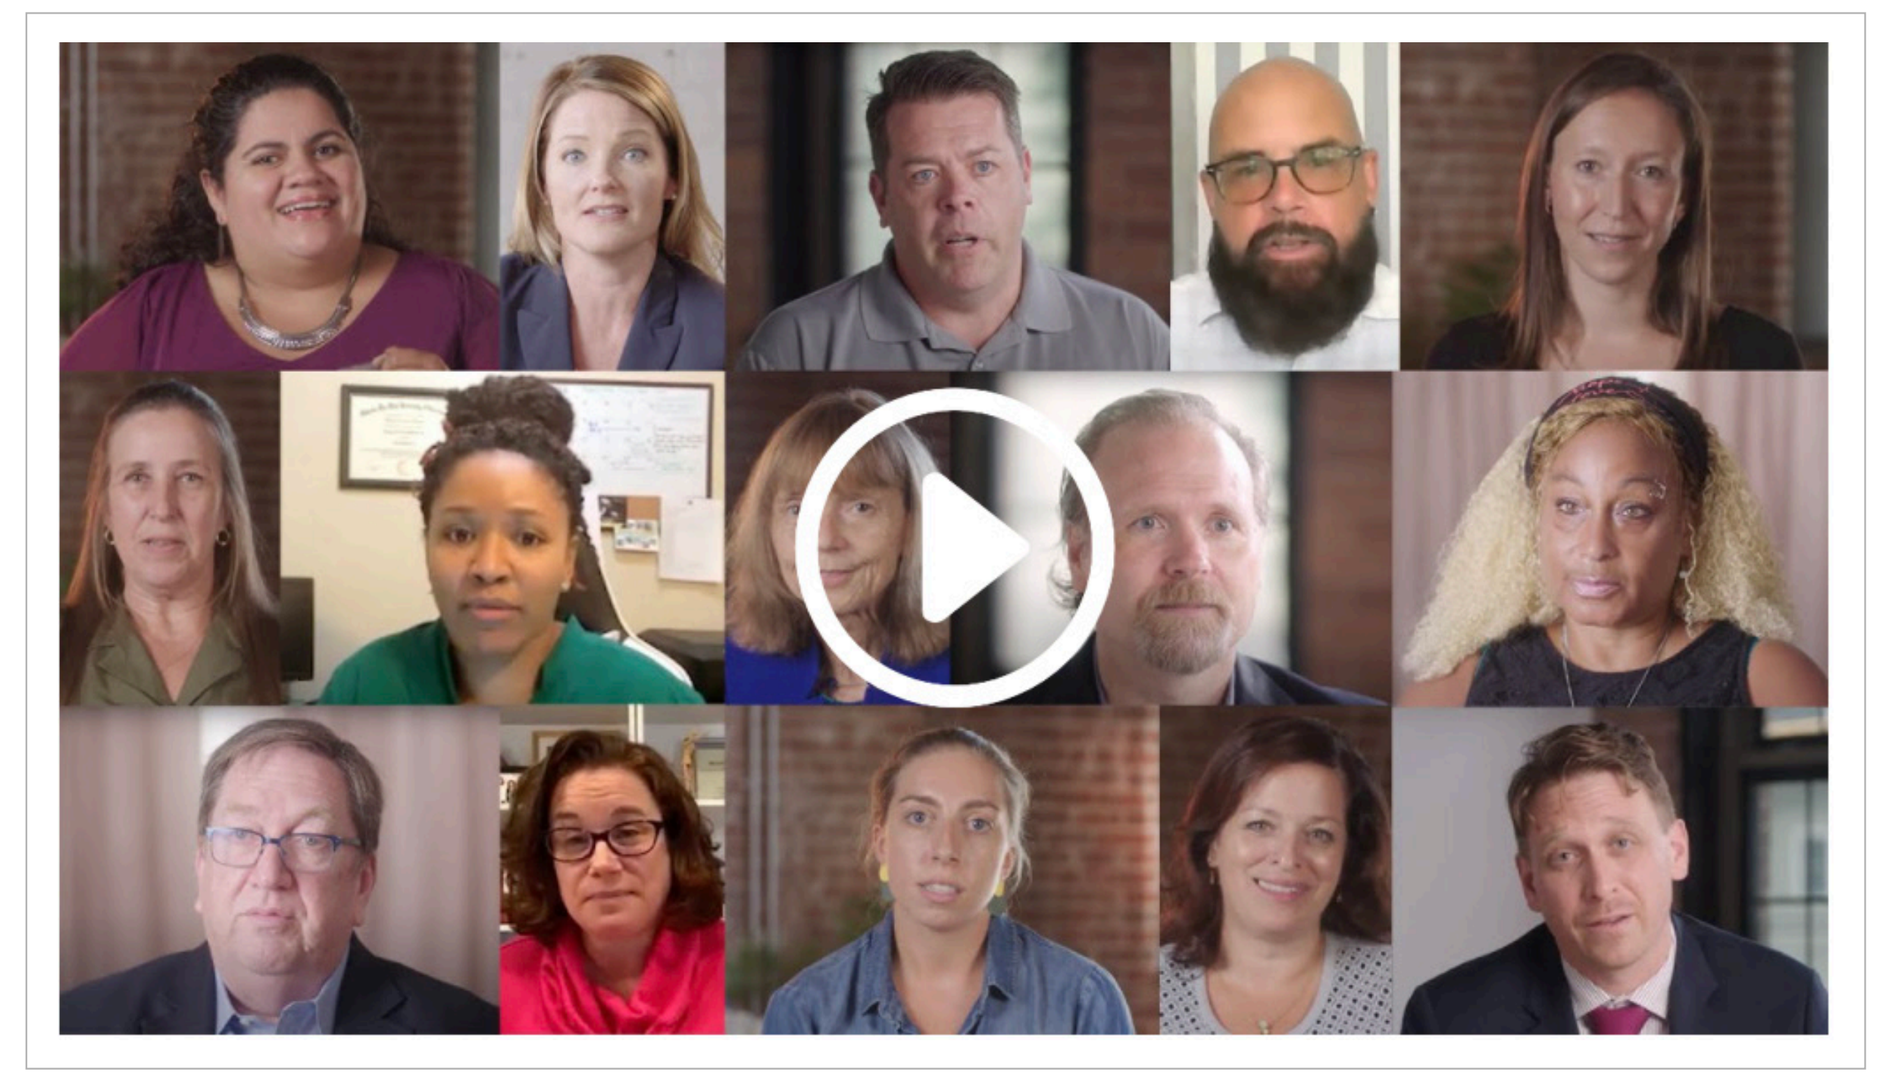

Toward a Unified Vision

SCAN ME

rize massachusetts
RIZE Massachusetts is the only public–private partnership solely dedicated to funding and creating solutions to end the overdose crisis. Guided by those with lived experience and unafraid of new ideas, RIZE is building networks, designing programming, and supporting community partners who are using novel approaches to preventing overdose. Since 2017, RIZE has granted over $8.6 million to community organizations in Massachusetts. www.rizema.org

C4 Innovations
C4 Innovations advances equitable access to recovery, wellness, and housing stability for people who are systematically marginalized. When all people—regardless of race, ethnicity, gender identity—are able to access and engage in quality care, the results are healthier individuals, families, and communities. www.c4innovates.com

LEARN to COPE
Learn to Cope is a peer-led support network that offers education, resources, and hope for family members and friends who have loved ones affected by substance use disorder. www.learn2cope.org

MOAR
MOAR, Massachusetts Organization for Addiction Recovery, is a statewide association of people in recovery, families, and friends educating the public about the value of addiction recovery since 1991. MOAR is a go-to organization for addiction recovery advocacy, education, and support services. MOAR believes that faces who are visible and voices who are vocal prove that supporting the path to recovery is valuable. That is how we become victorious! www.moar-recovery.org